Surveillance of Cefepime/enmetazobactam Against European Isolates of Enterobacterales Collected Between 2019 and 2021, Including Cephalosporin-resistant Phenotypes

P0289

Stephen Hawser[1*], Ian Morrissey[2], Nimmi Kothari[1], Federica Monti[1], Adam Belley[3], Nathalie Dunkel[4]

[1]IHMA Europe, Monthey, Switzerland; [2]Antimicrobial Focus, Sawbridgeworth, UK; [3]Allecra Therapeutics, Saint-Louis, France; [4]Advanz Pharma, London, UK

*Contact details: shawser@ihma.com

INTRODUCTION

Cefepime/enmetazobactam is a novel beta-lactam/beta-lactamase inhibitor combination intended as empiric therapy for serious infections proven or suspected to be caused by Gram-negative pathogens. In a phase 3 study of complicated urinary tract infections and acute pyelonephritis, cefepime/enmetazobactam met criteria for non-inferiority and superiority compared to piperacillin/tazobactam (1). The purpose of this study was to monitor the *in vitro* activity of cefepime/enmetazobactam against Enterobacterales surveillance isolates collected from European hospitals between 2019 and 2021, including isolates with phenotypic resistance to 3rd-generation cephalosporins (3GC).

MATERIALS AND METHODS

A total of 2,627 isolates were collected from Germany (DE), France (FR), the United Kingdom (UK), Italy (IT), Spain (ES), the Netherlands (NL), Belgium (BE), Sweden (SW) and Denmark (DK) which originated from a variety of infection sources (Figures 1 & 2).

Minimum inhibitory concentrations (MICs) were determined by Clinical and Laboratory Standards Institute (CLSI) broth microdilution methodology (2).

Cefepime/enmetazobactam was tested at a fixed concentration of 8 mg/L enmetazobactam. Antimicrobial susceptibility was determined using the 2022 European Committee on Antimicrobial Susceptibility Testing (EUCAST) breakpoints (3). Cefepime/enmetazobactam breakpoints are yet to be established, so the EUCAST cefepime breakpoints of susceptible ≤1 mg/L and susceptible-increased exposure ≤4 mg/L (assigned for a cefepime dose of 2 g every 8 hours) were used for comparative purposes.

Phenotypic resistance to 3GC was defined as resistant to ceftriaxone and ceftazidime but susceptible to the carbapenem meropenem.

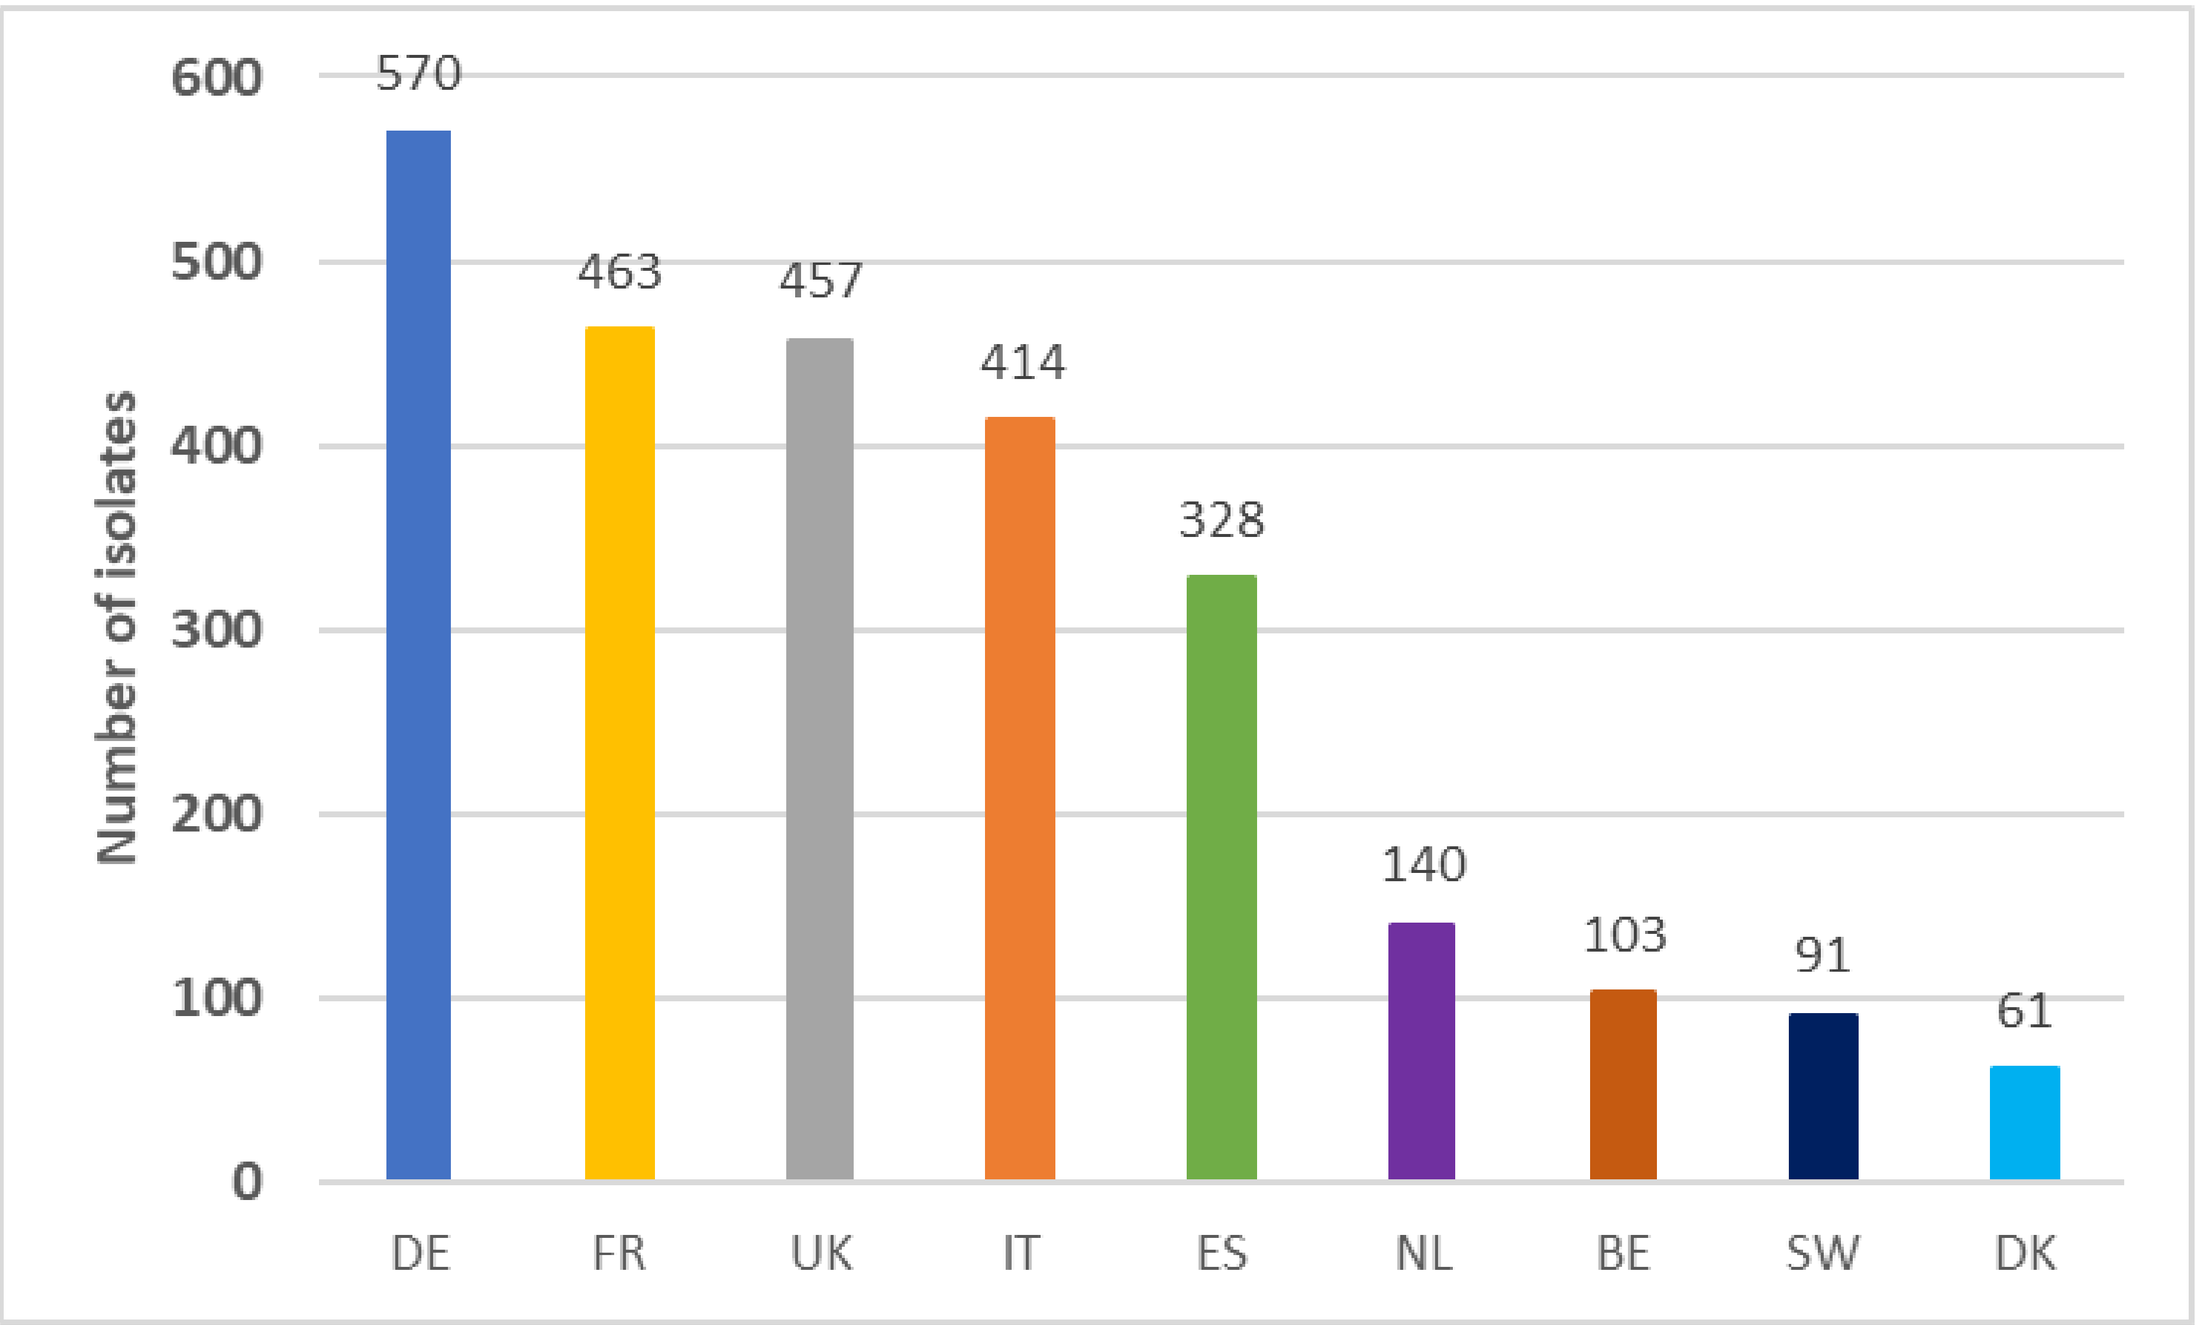
Number of isolates
600
500
400
300
200
100
0
570
463
457
414
328
140
103
91
61
DE
FR
UK
IT
ES
NL
BE
SW
DK

FIGURE 1: Isolate count by country of origin

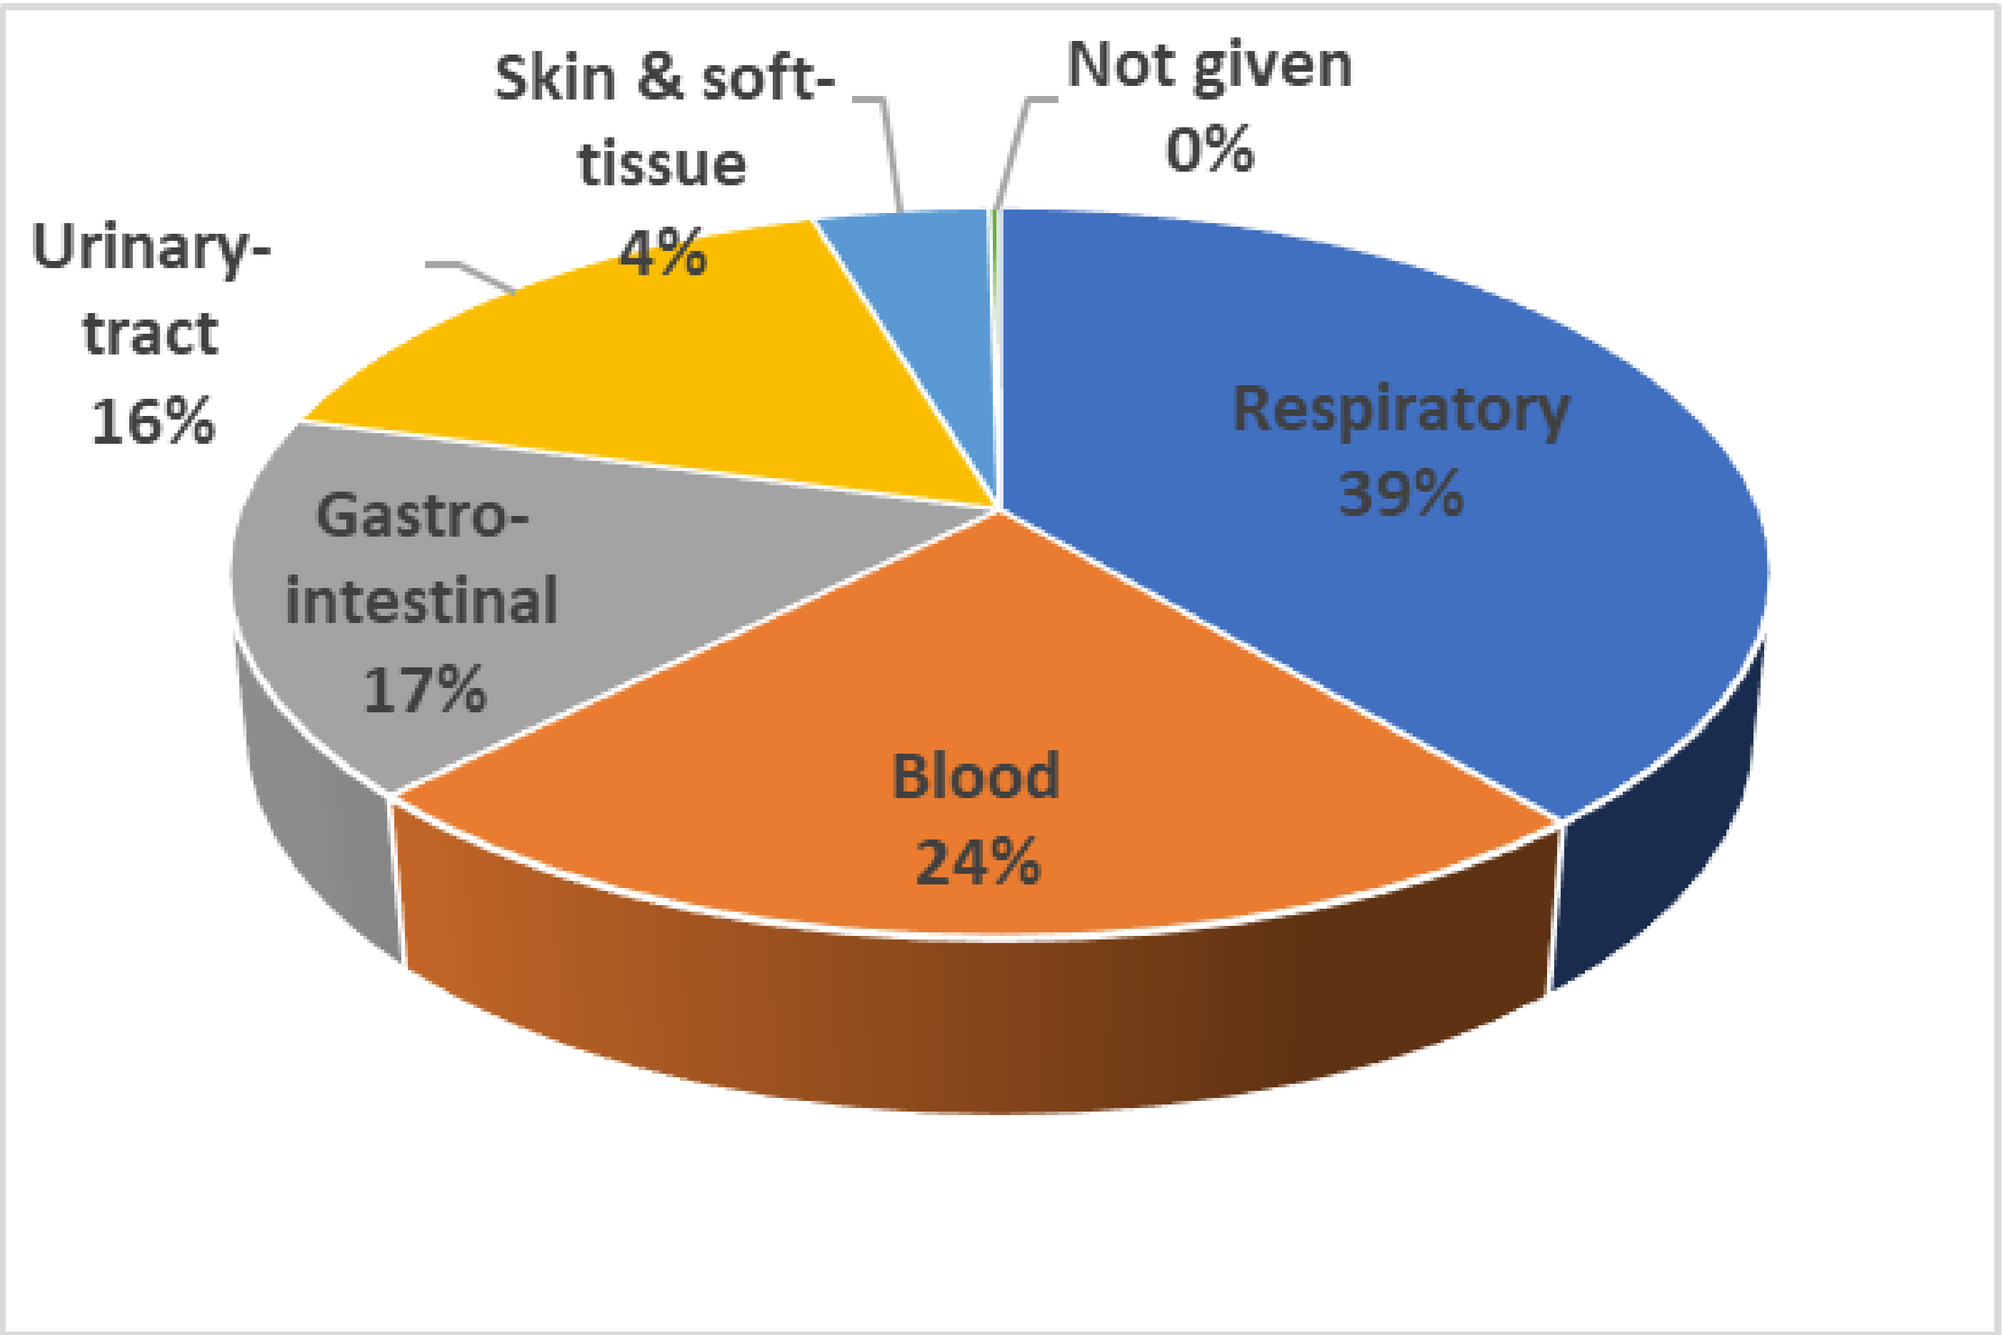
Skin & soft-tissue 4%
Not given 0%
Urinary-tract 16%
Respiratory 39%
Gastro-intestinal 17%
Blood 24%

FIGURE 2: Isolate infection source (source %)

RESULTS

TABLE 1: Summary of activity of cefepime/enmetazobactam and comparators against all Enterobacterales (n=2627)

Antimicrobial	MIC (mg/L):				Percentage:		
	Min	Max	50%	90%	Sus	Sus (IE)	Res
Cefepime	≤ 0.008	> 16	0.06	16	83.9	88.1	11.9
Cefepime/enmetazobactam (8 mg/L)	≤ 0.015	> 64	0.03	0.25	97.0	97.9	2.1
Ceftazidime	≤ 0.03	> 16	0.25	> 16	77.2	81.4	18.6
Ceftazidime/avibactam (4 mg/L)	≤ 0.12	> 16	≤ 0.12	0.5	99.4	-	0.6
Ceftriaxone	≤ 0.015	> 4	0.06	> 4	77.4	78.7	21.3
Ciprofloxacin	≤ 0.004	> 1	0.03	> 1	79.8	82.6	17.4
Meropenem	≤ 0.004	> 4	0.03	0.06	97.3	100.0	0.0
Piperacillin/tazobactam (4 mg/L)	≤ 0.5	> 128	2	64	80.8	-	19.2

Min/Max, minimum/maximum MIC; 50%/90%, concentration required to inhibit 50%/90% of isolates; Sus, susceptible; Sus (IE), susceptible (increased exposure); Res, resistant

TABLE 2: Summary of activity of cefepime/enmetazobactam and comparators against 3GC-resistant Enterobacterales (n=407)

Antimicrobial	MIC (mg/L):				Percentage:		
	Min	Max	50%	90%	Sus	Sus (IE)	Res
Cefepime	0.03	> 16	8	> 16	29.2	46.7	53.3
Cefepime/enmetazobactam (8 mg/L)	≤ 0.015	32	0.12	1	96.3	99.3	0.7
Ceftazidime	8	> 16	> 16	> 16	0.0	0.0	100.0
Ceftazidime/avibactam (4 mg/L)	≤ 0.12	> 16	0.25	1	99.5	99.5	0.5
Ceftriaxone	4	> 4	> 4	> 4	0.0	0.0	100.0
Ciprofloxacin	≤ 0.004	> 1	0.5	> 1	44.0	51.6	48.4
Meropenem	0.015	2	0.03	0.12	100.0	100.0	0.0
Piperacillin/tazobactam (4 mg/L)	≤ 0.5	> 128	16	128	39.6	-	60.4

Min/Max, minimum/maximum MIC; 50%/90%, concentration required to inhibit 50%/90% of isolates; Sus, susceptible; Sus (IE), susceptible (increased exposure); Res, resistant

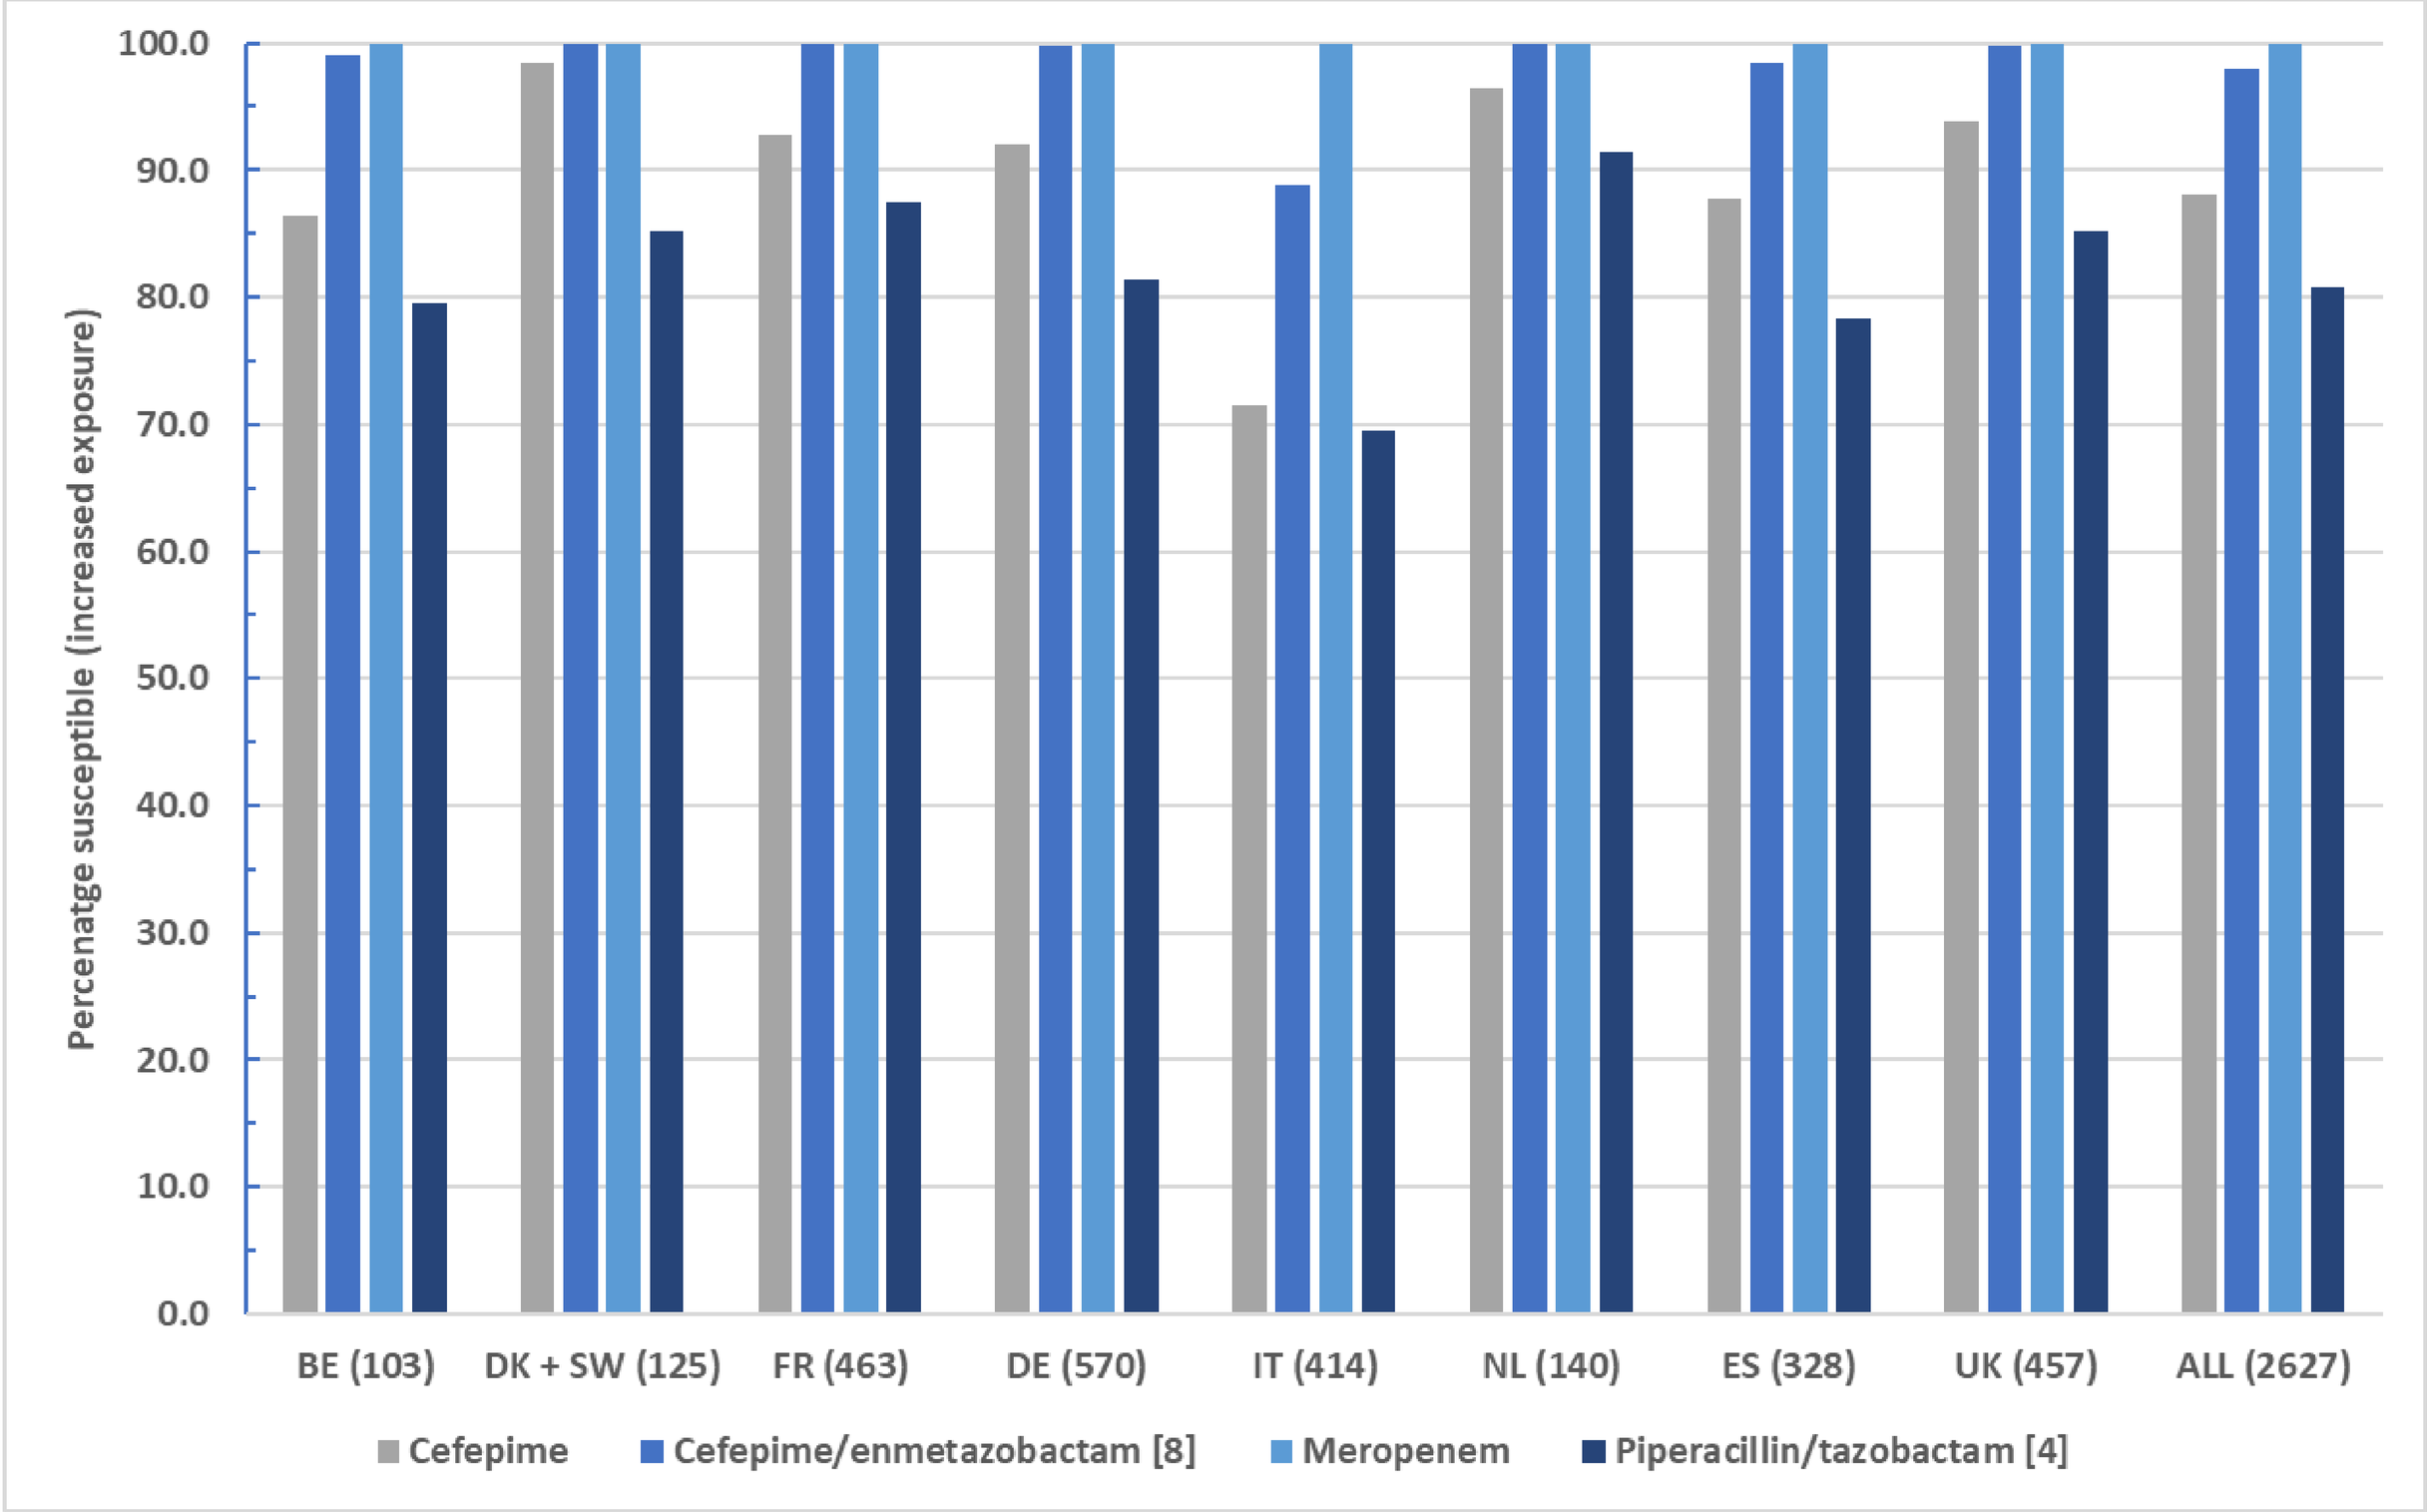
Percenatge susceptible (increased exposure)
BE (103)
DK + SW (125)
FR (463)
DE (570)
IT (414)
NL (140)
ES (328)
UK (457)
ALL (2627)
Cefepime
Cefepime/enmetazobactam [8]
Meropenem
Piperacillin/tazobactam [4]

FIGURE 3: Susceptibility of Enterobacterales isolates by country (number of isolates) to cefepime/enmetazobactam and select comparators

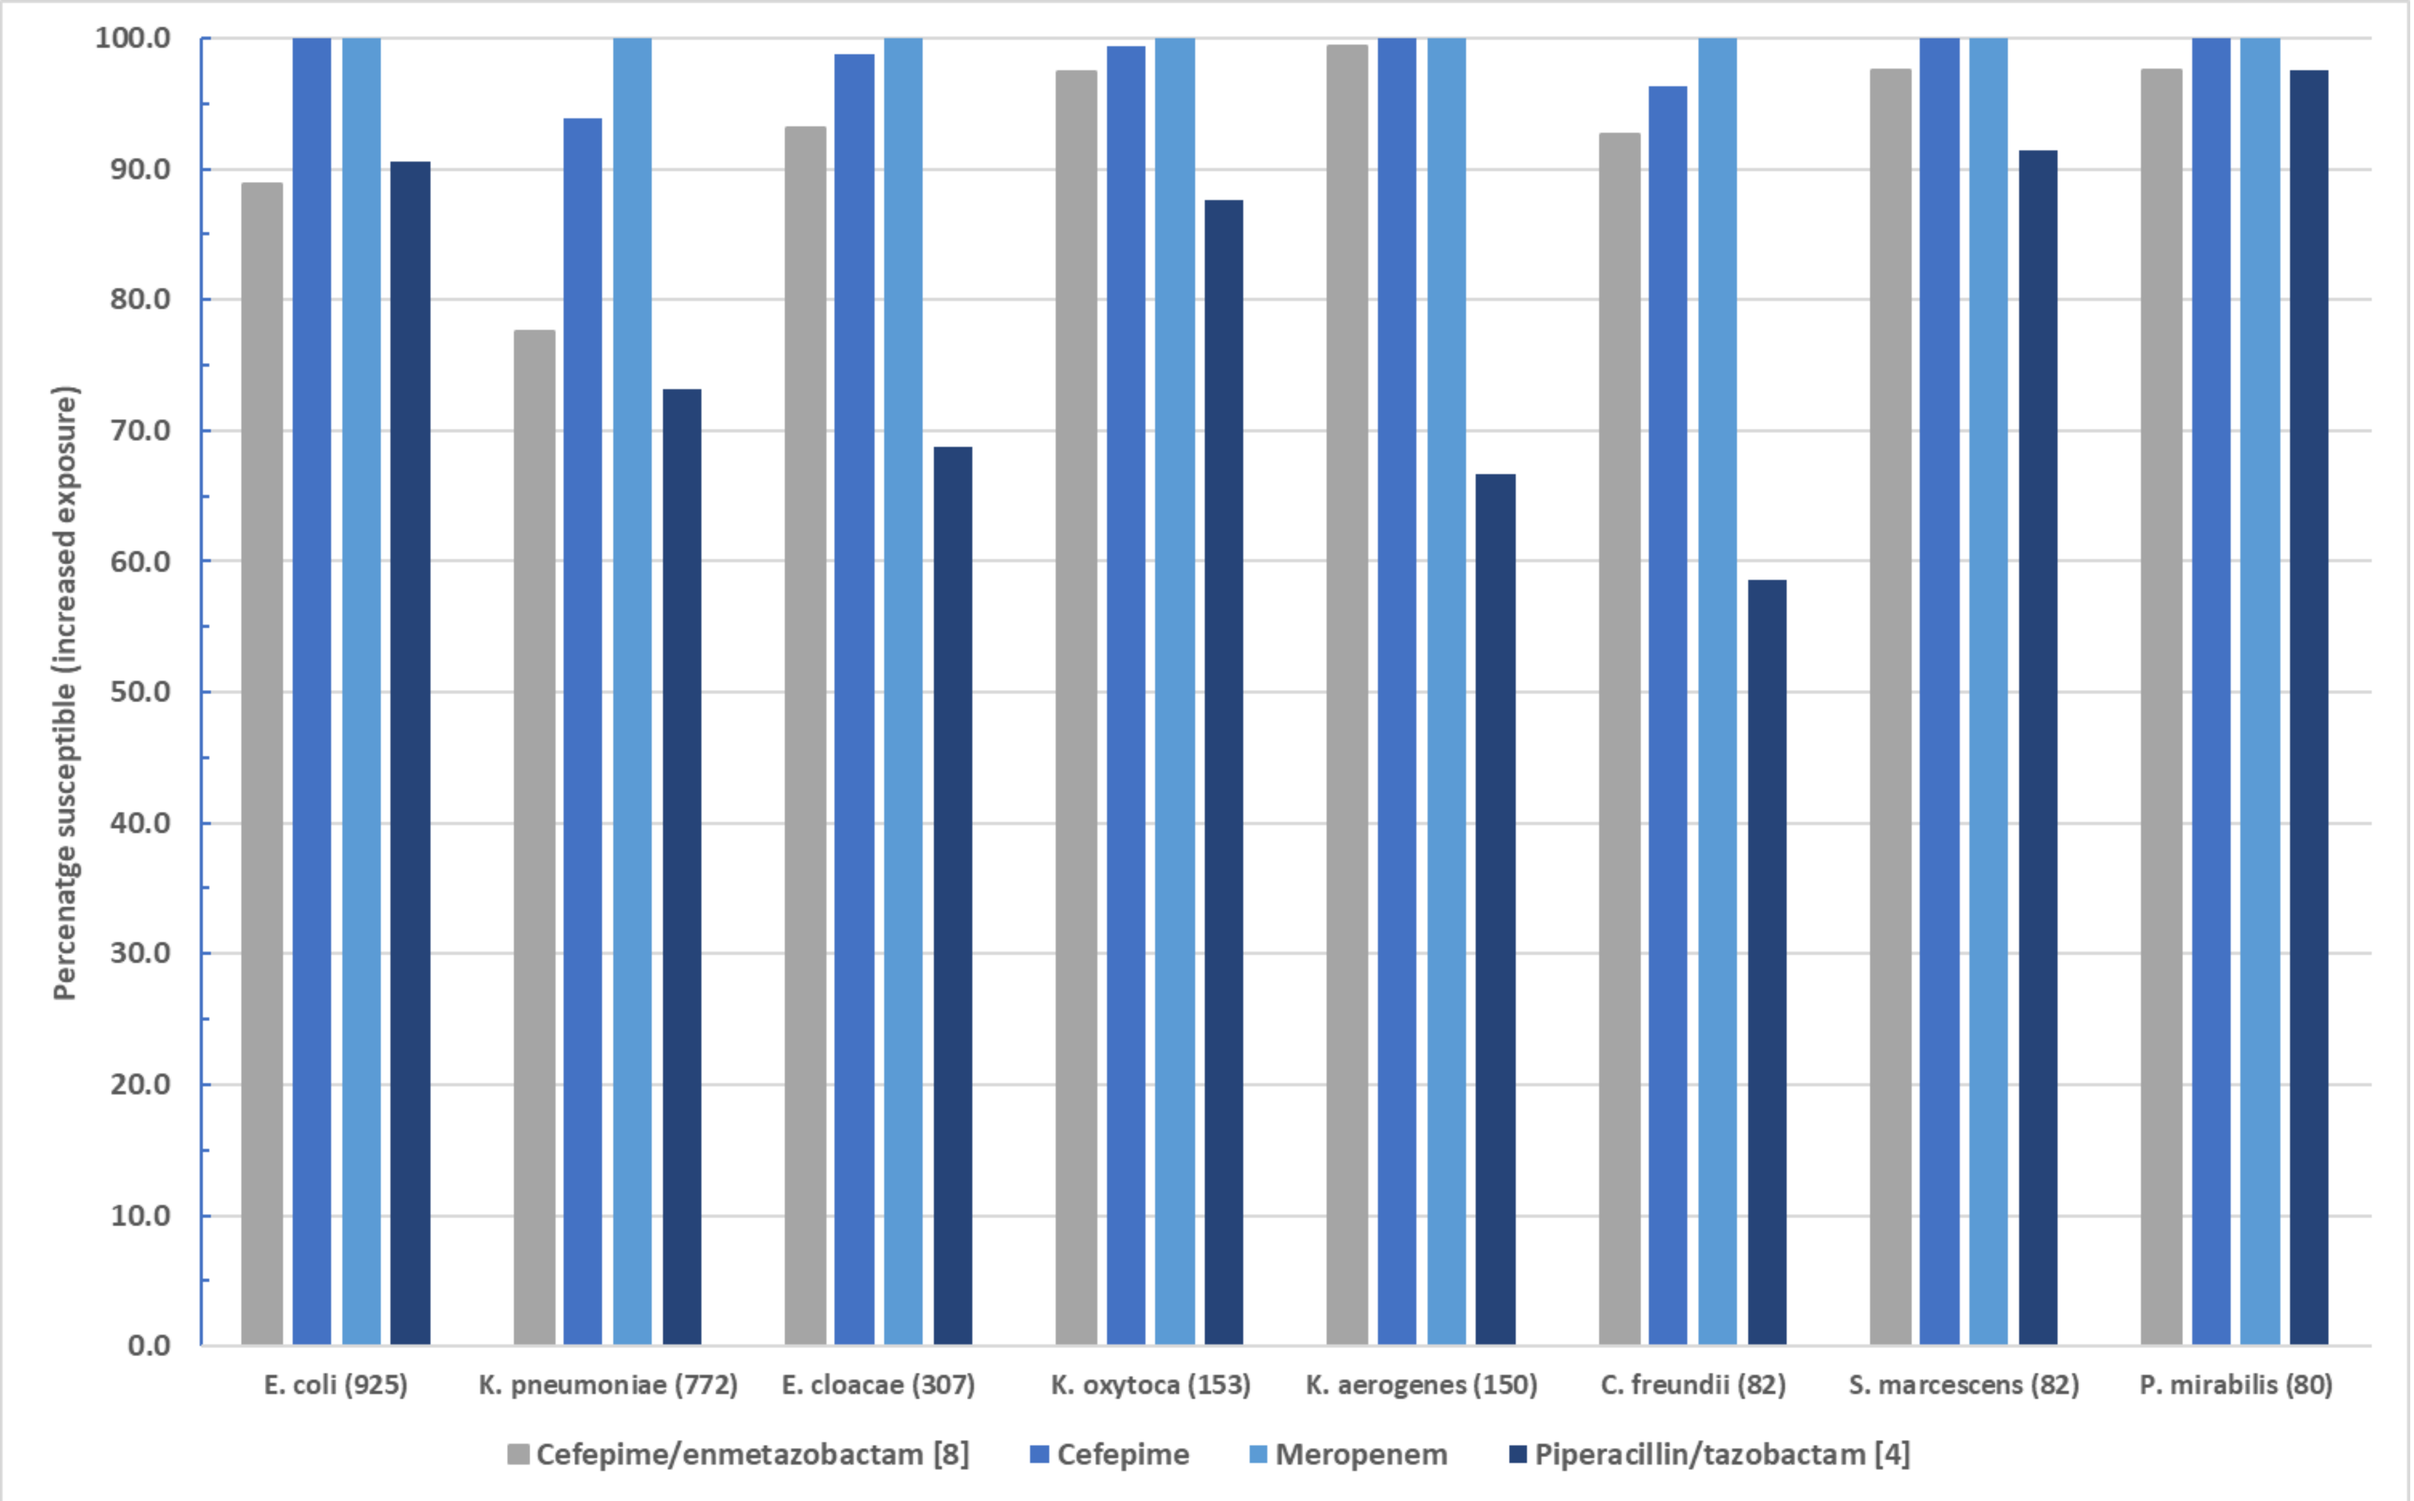
Percenatge susceptible (increased exposure)
E. coli (925)
K. pneumoniae (772)
E. cloacae (307)
K. oxytoca (153)
K. aerogenes (150)
C. freundii (82)
S. marcescens (82)
P. mirabilis (80)
Cefepime/enmetazobactam [8]
Cefepime
Meropenem
Piperacillin/tazobactam [4]

FIGURE 4: Susceptibility of individual Enterobacterales species (number of isolates) to cefepime/enmetazobactam and select comparators

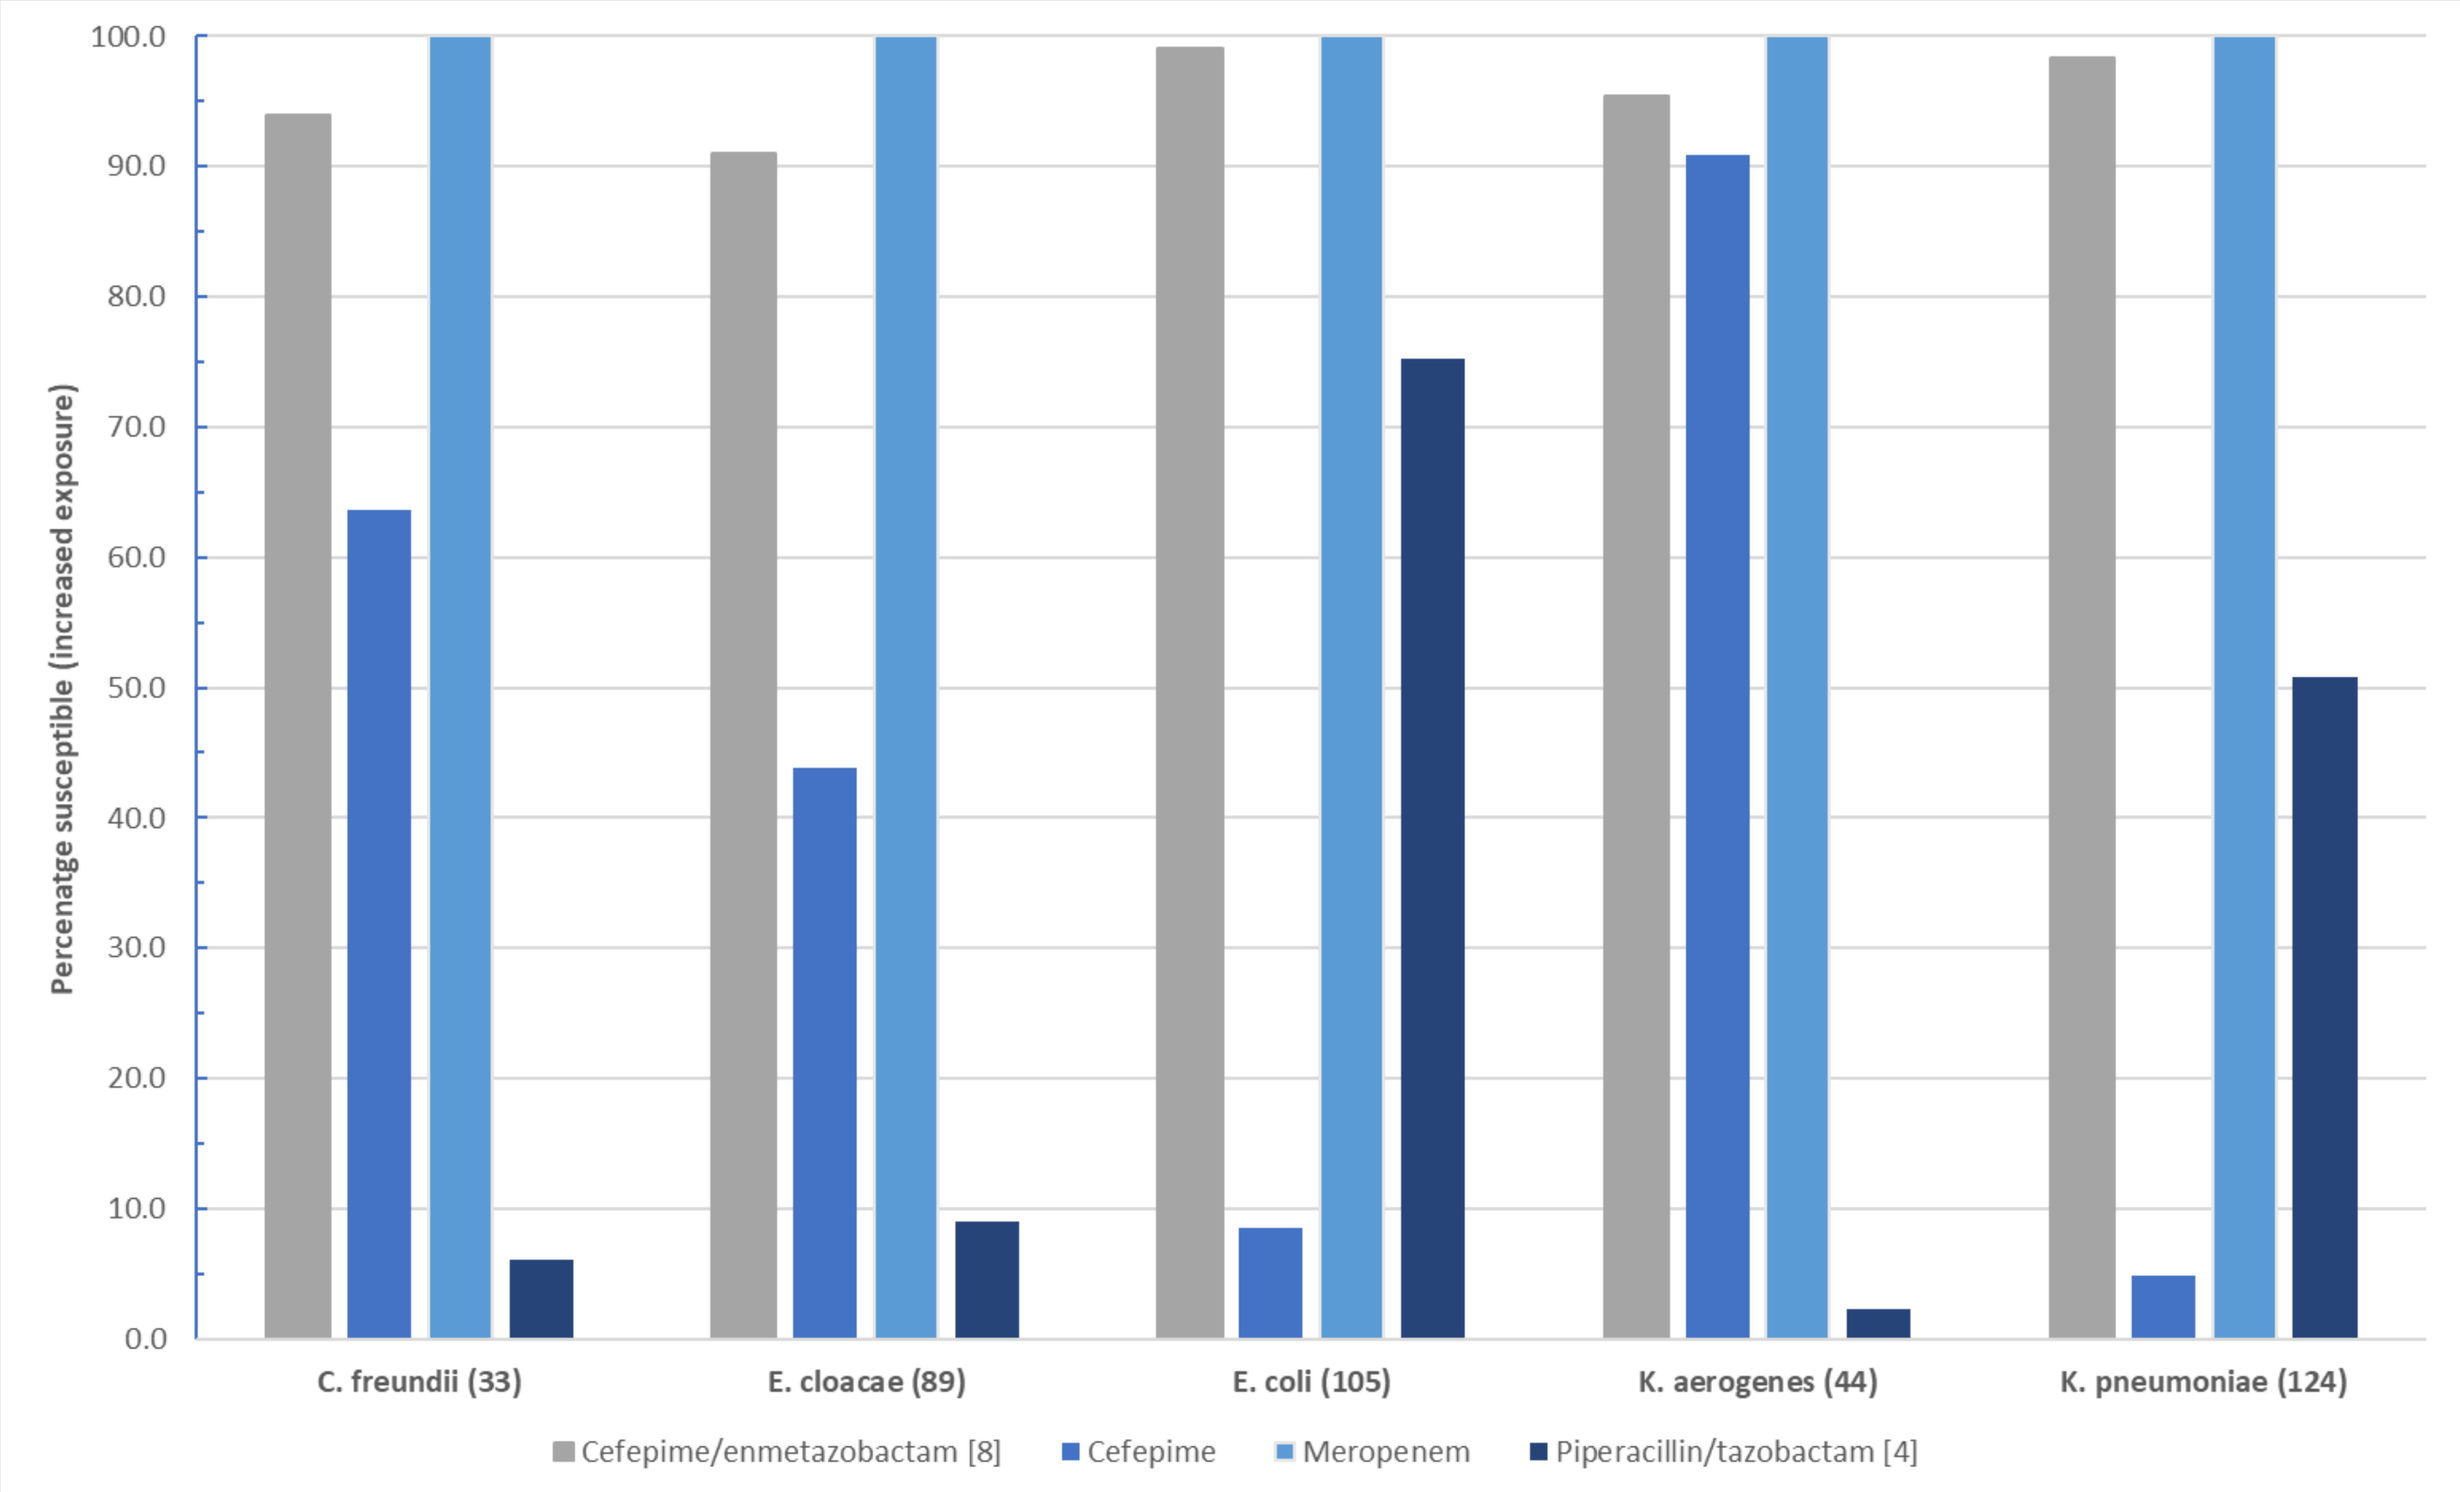
Percenatge susceptible (increased exposure)
C. freundii (33)
E. cloacae (89)
E. coli (105)
K. aerogenes (44)
K. pneumoniae (124)
Cefepime/enmetazobactam [8]
Cefepime
Meropenem
Piperacillin/tazobactam [4]

FIGURE 5: Susceptibility of 3GC-resistant Enterobacterales species (number of isolates) to cefepime/enmetazobactam and select comparators

RESULTS SUMMARY

- Susceptibility of Enterobacterales isolates to cefepime/enmetazobactam, using provisional breakpoints, was 97.0/97.9% overall. This ranged from 87.2% in Italy to 100% in Denmark and the Netherlands by country and 92.6% in *K. pneumoniae* to 100% in *P. mirabilis*, *P. rettgeri* and *P. stuartii* by species.
- Cephalosporin and piperacillin-tazobactam susceptibility overall was lower, ranging from 77.2% (ceftazidime) to 83.9% (cefepime).
- 3GC-resistance was 15.5% (407/2627) overall with only 39.6% susceptibility to piperacillin/tazobactam versus 96.3/99.3% susceptibility to cefepime/enmetazobactam, which was comparable to meropenem.

REFERENCES

1. Kaye KS et al. 2022. Effect of Cefepime/Enmetazobactam vs Piperacillin/Tazobactam on Clinical Cure and Microbiological Eradication in Patients With Complicated Urinary Tract Infection or Acute Pyelonephritis: A Randomized Clinical Trial. *JAMA* **328**:1304-14.
2. CLSI. 2018. Methods for Dilution Antimicrobial Susceptibility Tests for Bacteria That Grow Aerobically. 11th ed. CLSI standard M07. CLSI, Wayne, PA, USA.
3. EUCAST. 2022. Breakpoint tables for interpretation of MICs and zone diameters. Version 12.0. http://www.eucast.org

CONCLUSIONS

- The combination of the cefepime/enmetazobactam retains potent *in vitro* activity against contemporary European clinical isolates of Gram-negative pathogens including those exhibiting phenotypic resistance to 3rd-generation cephalosporins.
- These data demonstrate the erosion of susceptibility to cephalosporins and other important antibiotic classes due to antibiotic resistance.

Presented at the 33rd European Congress of Clinical Microbiology & Infectious Diseases, Copenhagen, Denmark on 15 - 18 April 2023